ERS | Alpha-1 Antitrypsin Conference 2026

23–24 October 2026 | Barcelona, Spain

In collaboration with EARCO European Alpha-1 Research Collaboration

#ERSAlpha12026

CO-8195, a novel orally active small-molecule corrector for Alpha-1 Antitrypsin Deficiency (AATD), increases functional secreted Z-A1AT and reduces polymerized Z-A1AT in cellular assays and in the PiZ mouse model

Anzhelika Kabro, Nathalia Azevedo Portilho, Édith Bellavance, Claire Bonfils, Alex Caron, Lee Fader, Anelya Gandy, Michael R. Goldsmith, Liying Gong, Steven Moore, Shivani Patel, Paulina Wojnarowicz, Sharath Hegde

Congruence Therapeutics Inc., Montréal, Canada, www.congruencetx.com

Contact info: akabro@congruencetx.com

Goal, discovery and mechanism of action

Congruence goal

- To develop a *differentiated*, **optimized oral small-molecule corrector**
- with the potential to restore **protective levels of functional A1AT**
- **liver-targeted**: achieving optimal hepatic exposure and effectively **reduce Z-A1AT polymer burden**
- To address **both lung and liver disease**

Small-molecule Corrector

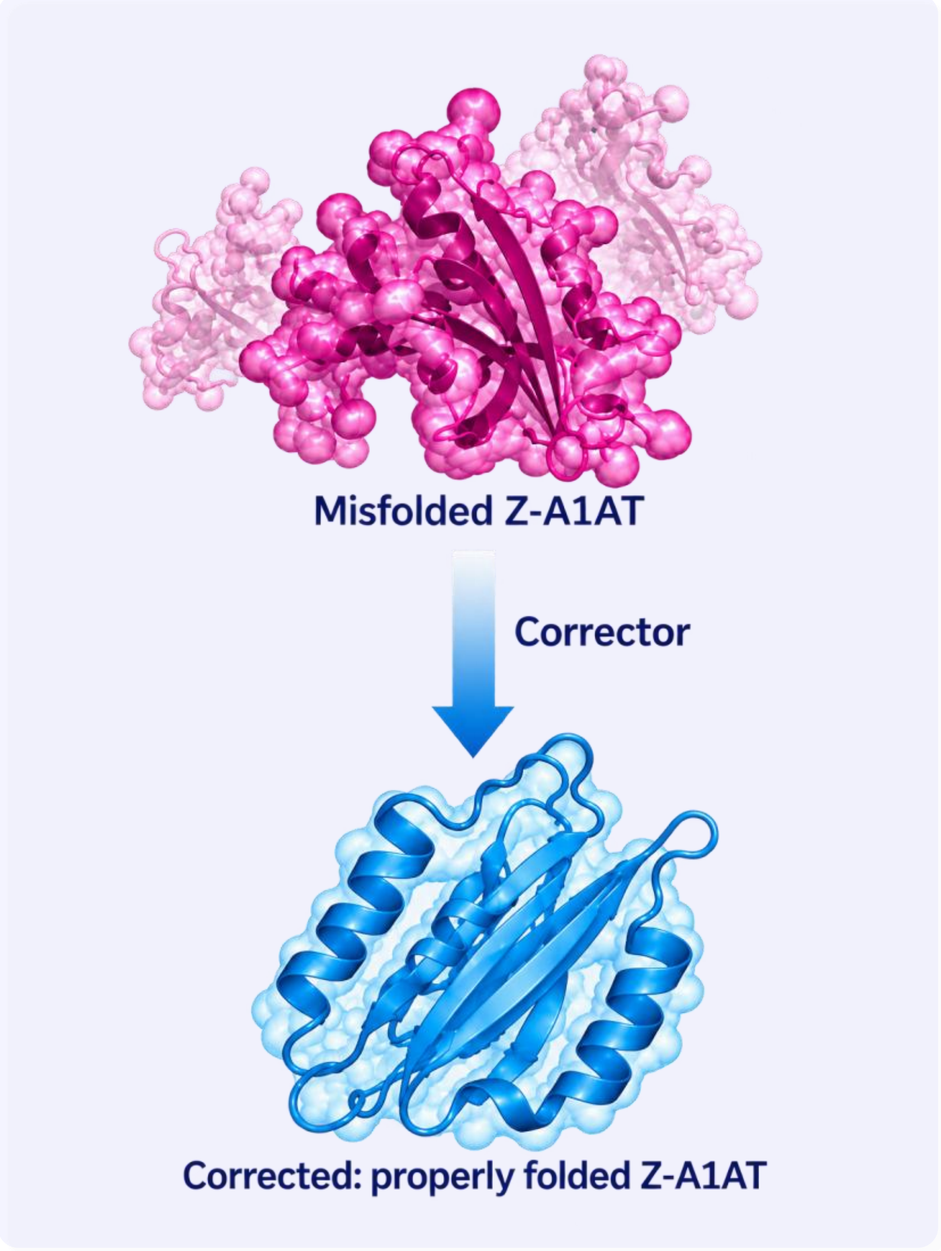
Misfolded Z-A1AT
Corrector
Corrected: properly folded Z-A1AT

Revenir™ engine

- The Revenir™ computational engine identifies pockets where small molecules may restore protein function and predicts small-molecule hits
- **CO-8195** reverses conformational landscape from Z-mutant → wild-type (WT) state

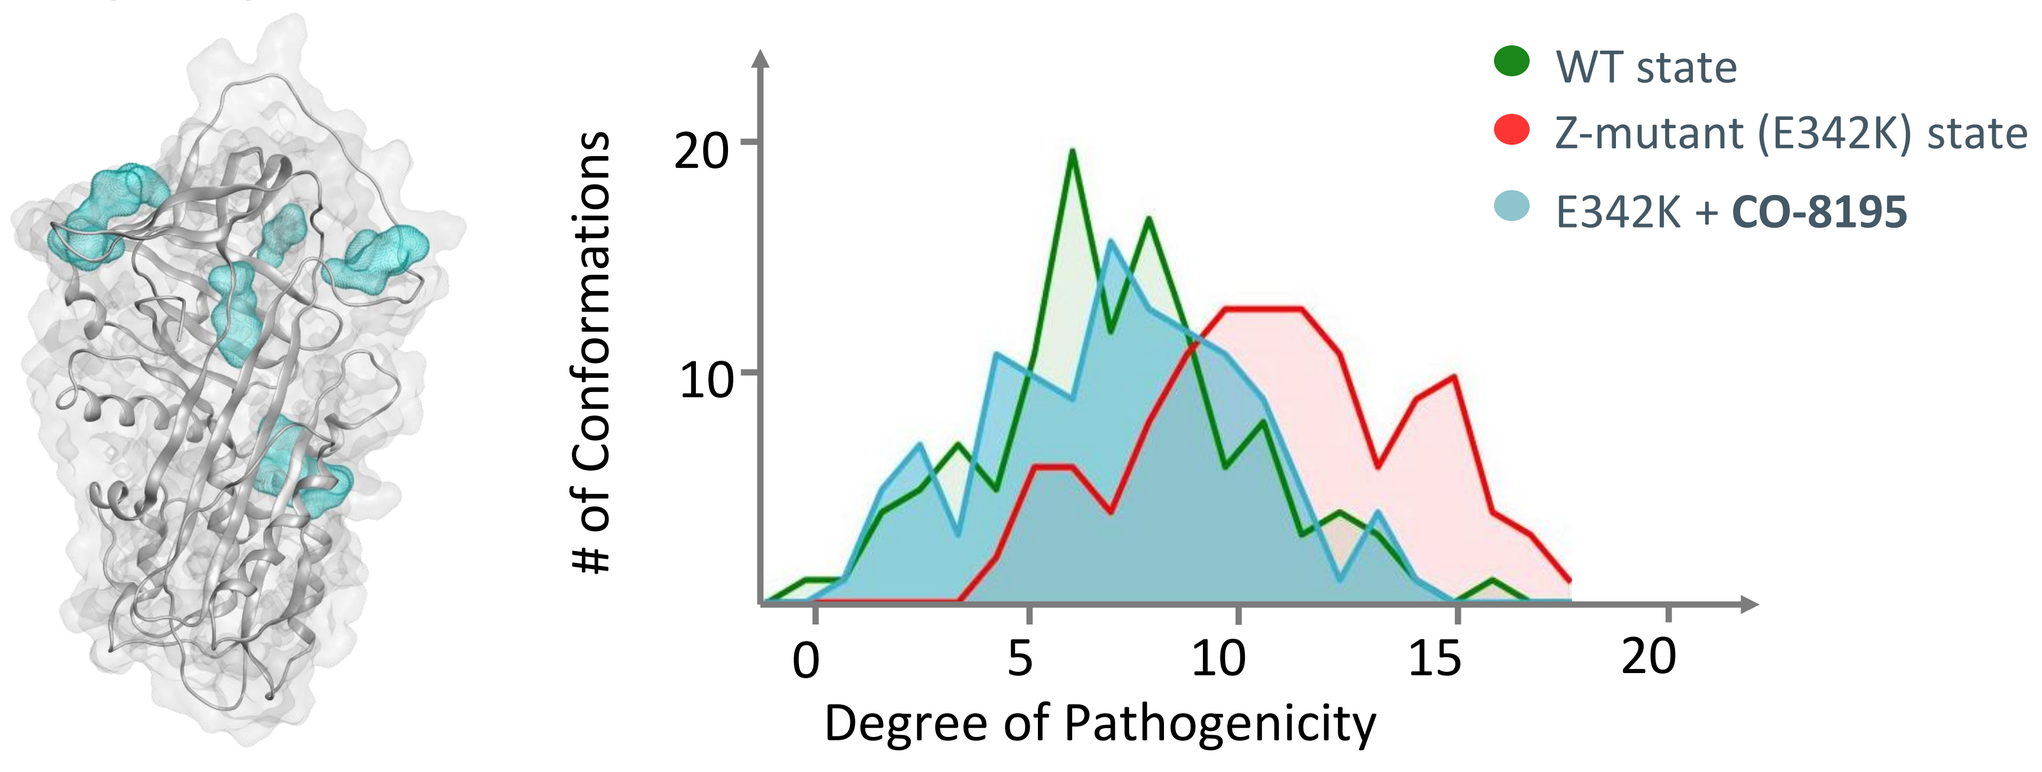
WT state
Z-mutant (E342K) state
E342K + CO-8195
of Conformations
20
10
0
5
10
15
20
Degree of Pathogenicity

CO-8195 Mechanism of Action

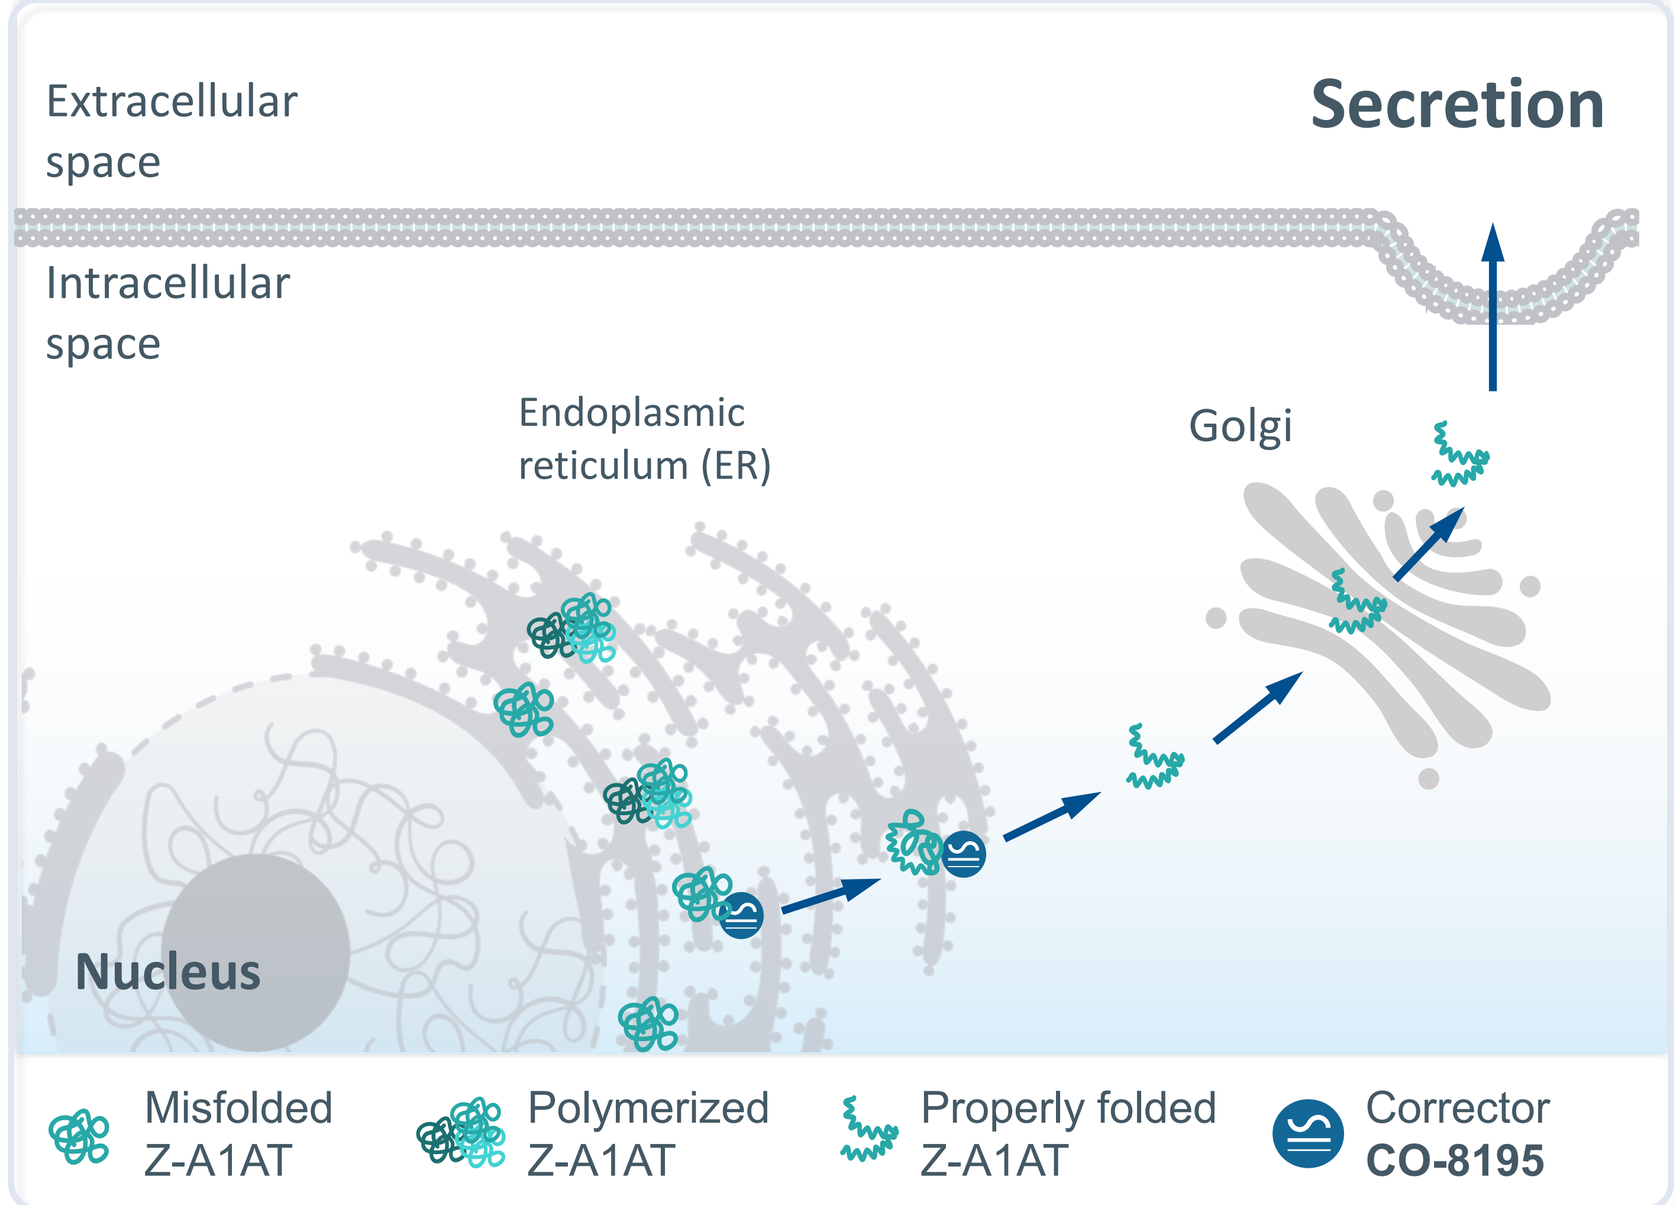
Extracellular space
Secretion
Intracellular space
Endoplasmic reticulum (ER)
Golgi
Nucleus
Misfolded Z-A1AT
Polymerized Z-A1AT
Properly folded Z-A1AT
Corrector CO-8195

In Vitro Characterization of CO-8195 Effects

Concentration-dependent ↑ Z-A1AT secretion, ↓ Z-A1AT polymer levels and Z-A1AT *h*NE inhibition function restauration

Assay	Z-A1AT Secretion	Z-A1AT Function	Z-A1AT Polymer Reduction
Method	ELISA	Enzymatic	In-cell Western
Sample type	Cell supernatant	Cell supernatant	Fixed cells
Parameter analyzed	Z-A1AT in cell supernatant (A1AT antibody)	Activity of *h*NE in presence of *h*NE substrate	Z-A1AT polymer (2C1 antibody)
Evaluated in target-relevant, human liver-derived **HepG2 cells** homozygous for E342K (Z-allele)	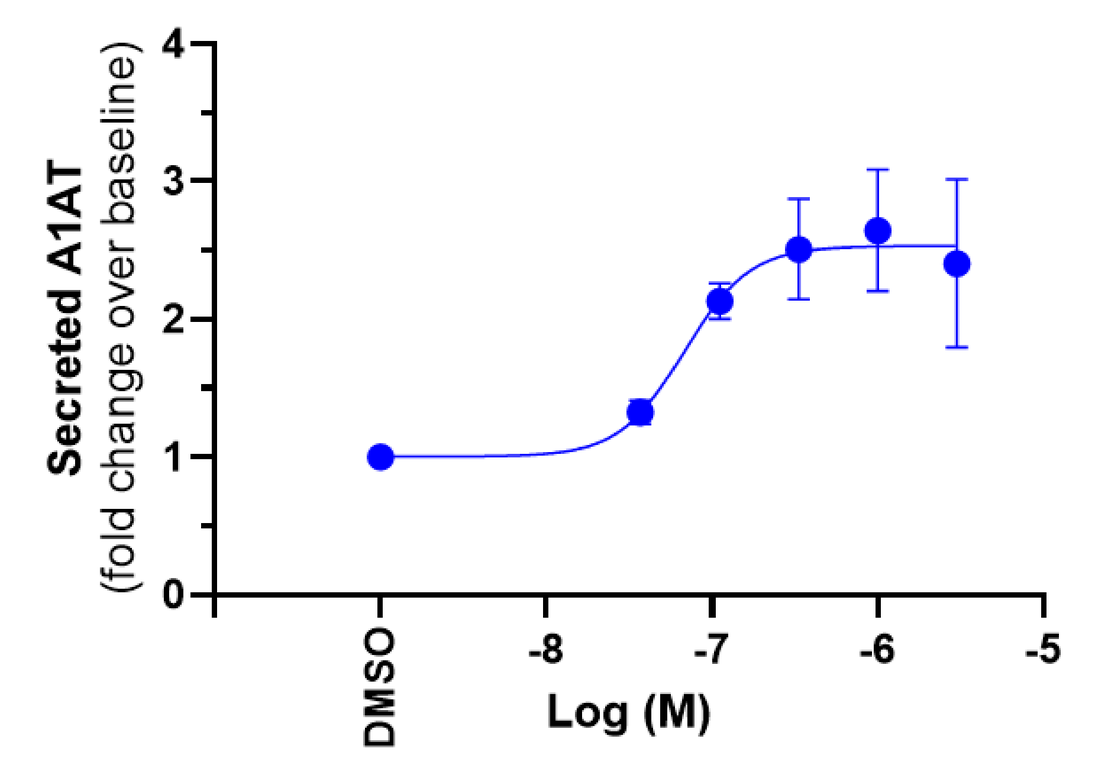 Secreted A1AT (fold change over baseline) 4 3 2 1 0 DMSO -8 -7 -6 -5 Log (M)	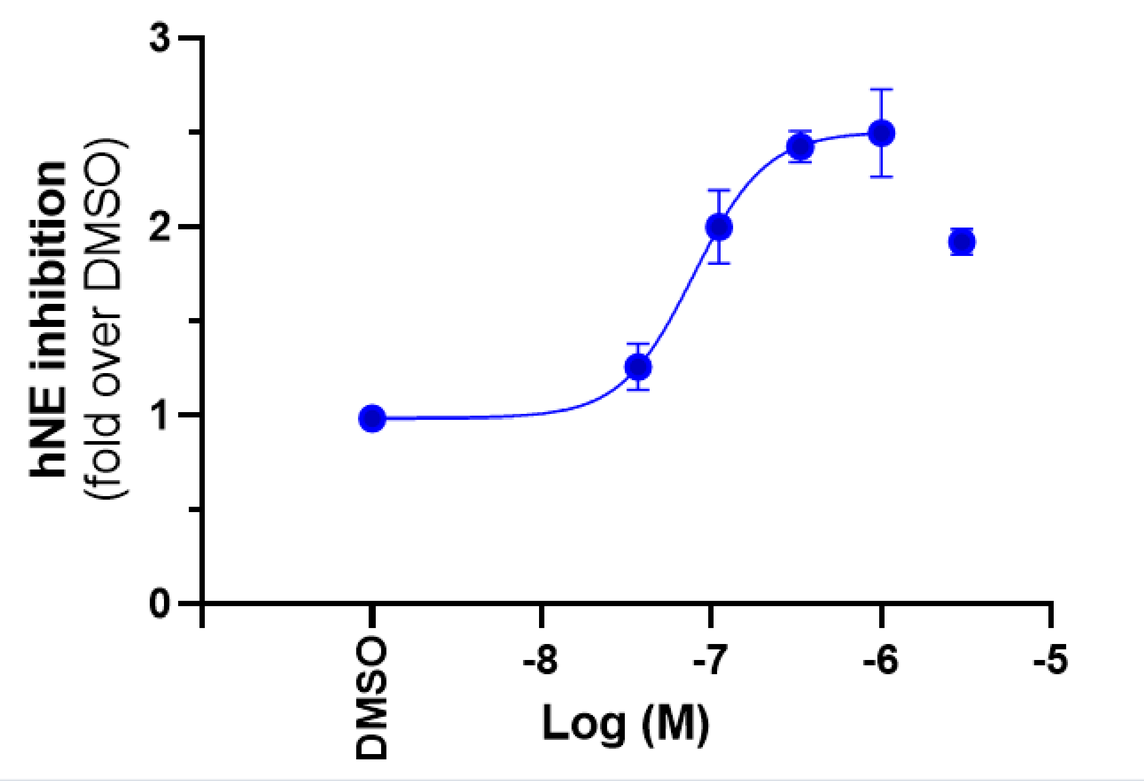 hNE inhibition (fold over DMSO) 3 2 1 0 DMSO -8 -7 -6 -5 Log (M)	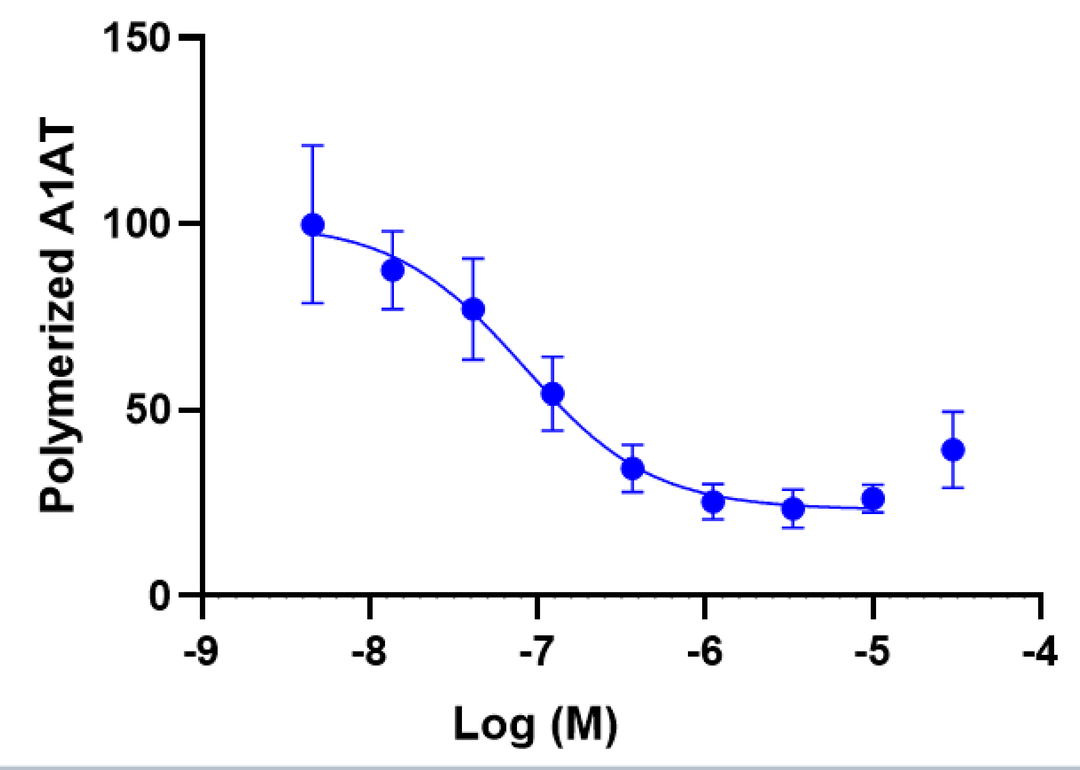 Polymerized A1AT 150 100 50 0 -9 -8 -7 -6 -5 -4 Log (M)
EC_{50} (95% CI, nM) / E_{max} (±SD)	**68 nM** (4.0-116.8) / **2.6-fold** (±0.44)	**79 nM** (60.3-102.3)/ **2.5-fold** (±0.23)	**82 nM** (18.4-152.5)

ADMET and *in Vivo* Evaluation

Excellent ADME properties

Orally bioavailable with preferential liver distribution

- ✓ High oral bioavailability (%F)
- ✓ Low-to-moderate clearance (CL)
- ✓ High liver-to-plasma ratio (Kp ~ 7) in preclinical species and PiZ mice

Liver Distribution (Kp) assessment

Setting	Tissue	Species	Kp
In vitro	Hepatocytes	*mouse*	**7.5**
		human	**3.6**
In vivo	Liver	*WT mouse*	**6.8**
		PiZ mouse	**7.7**

Non mutagenic (AMES test negative)

Favorable hERG profile

12-Week Oral Therapy in PiZ Mice: CO-8195 Produces Durable Efficacy

PiZ mouse model

- Strain #035411 (JAX)
- Carries ~16 human Z-A1AT copies
- Age at study start: 5-8 weeks, ♂

Pharmacodynamic endpoints:

- Restoration of plasma Z-A1AT function (binding to neutrophil elastase)
- Reduction of systemic Z-A1AT polymers in plasma
- Reduction of liver Z-A1AT polymers (by ELISA, histology)

Functional Z-A1AT in plasma

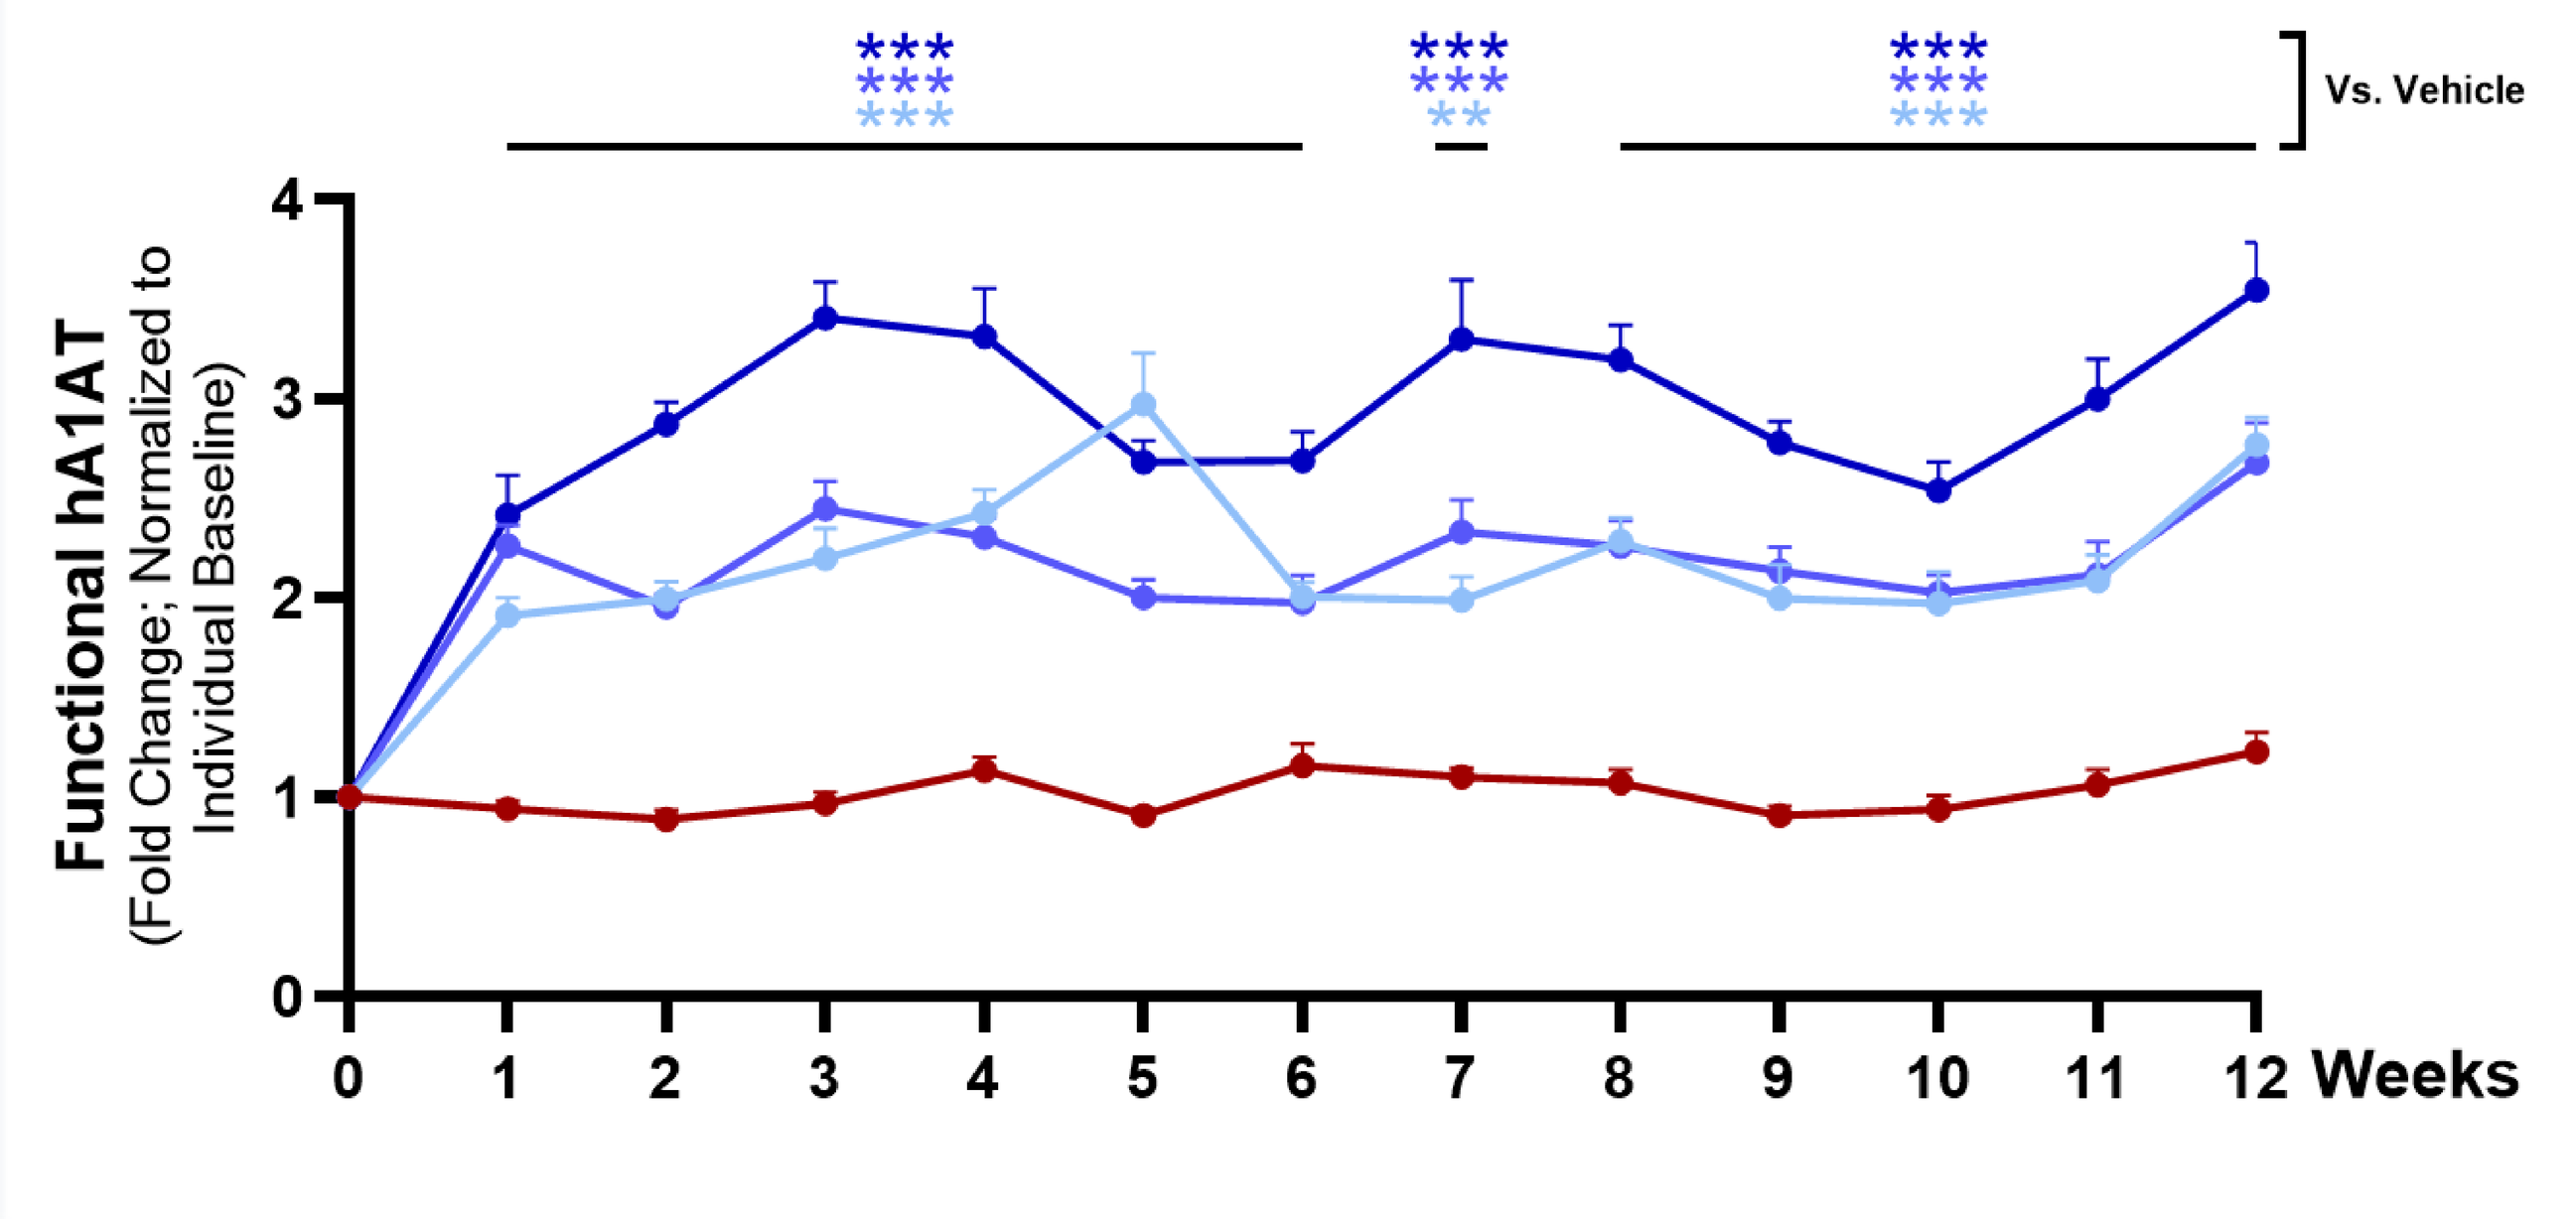
*** *** ***
*** *** **
*** *** ***
Vs. Vehicle
Functional hA1AT (Fold Change; Normalized to Individual Baseline)
4
3
2
1
0
0 1 2 3 4 5 6 7 8 9 10 11 12 Weeks

Polymer Z-A1AT in plasma

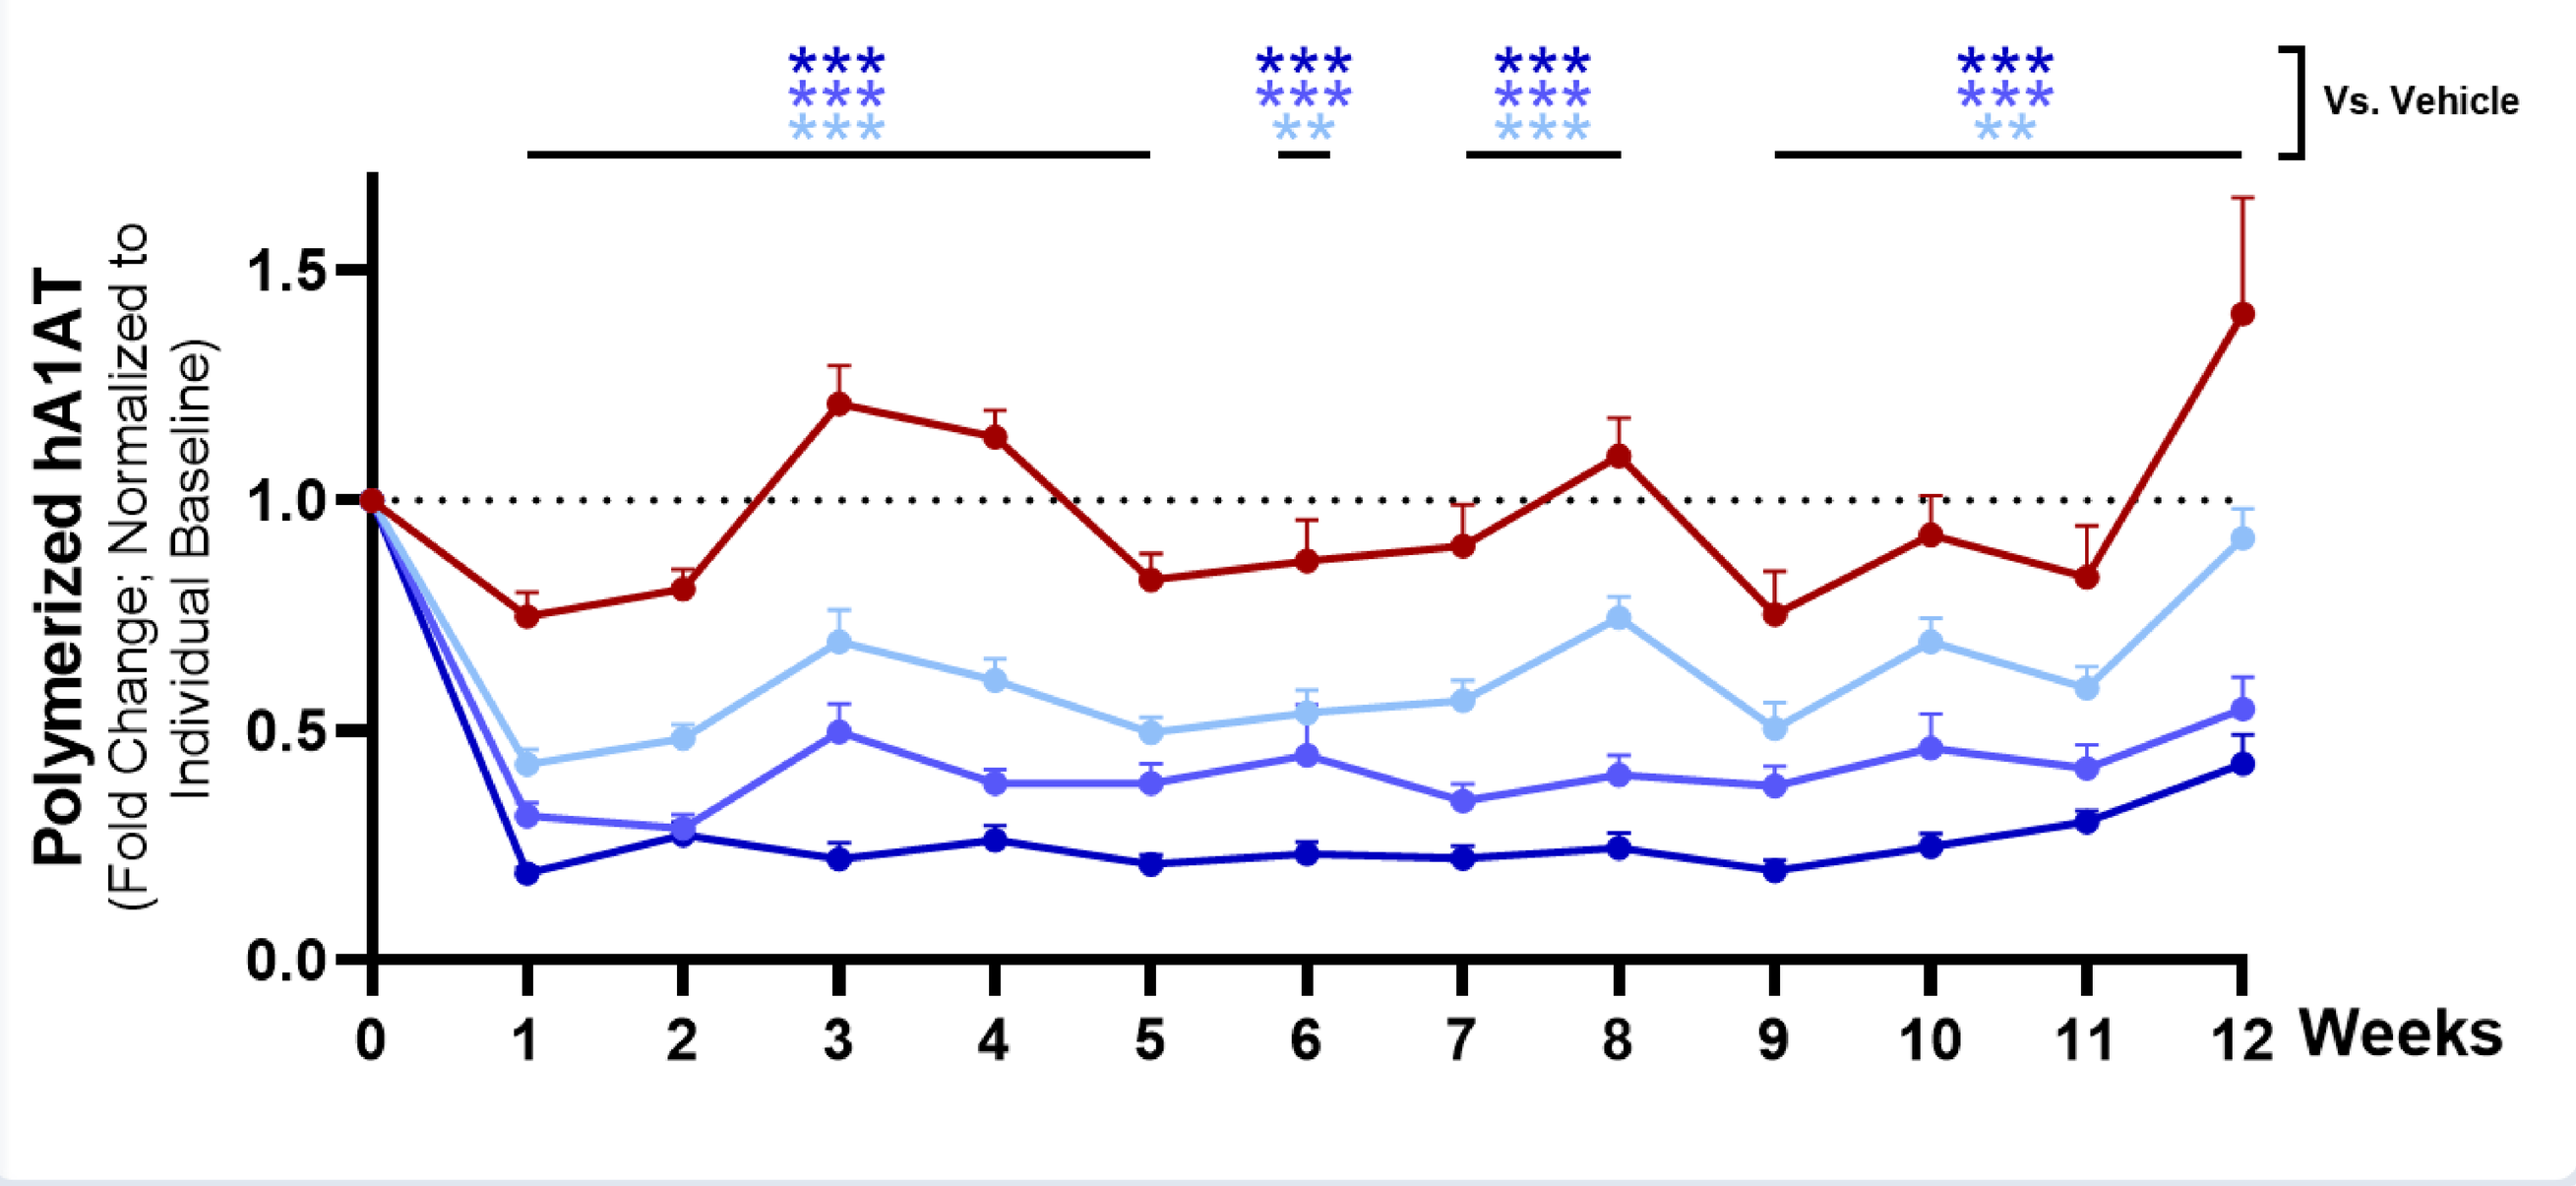
*** *** *** ***
*** *** *** ***
*** ** *** **
Vs. Vehicle
Polymerized hA1AT (Fold Change; Normalized to Individual Baseline)
1.5
1.0
0.5
0.0
0 1 2 3 4 5 6 7 8 9 10 11 12 Weeks

Vehicle (red); CO-8195: 30, 60, 120 mg/kg PO QD (light to dark blue).

Liver efficacy in PiZ Mice after 12-Week Oral Therapy: CO-8195 Induces Liver Polymer Clearance

Vehicle-treated

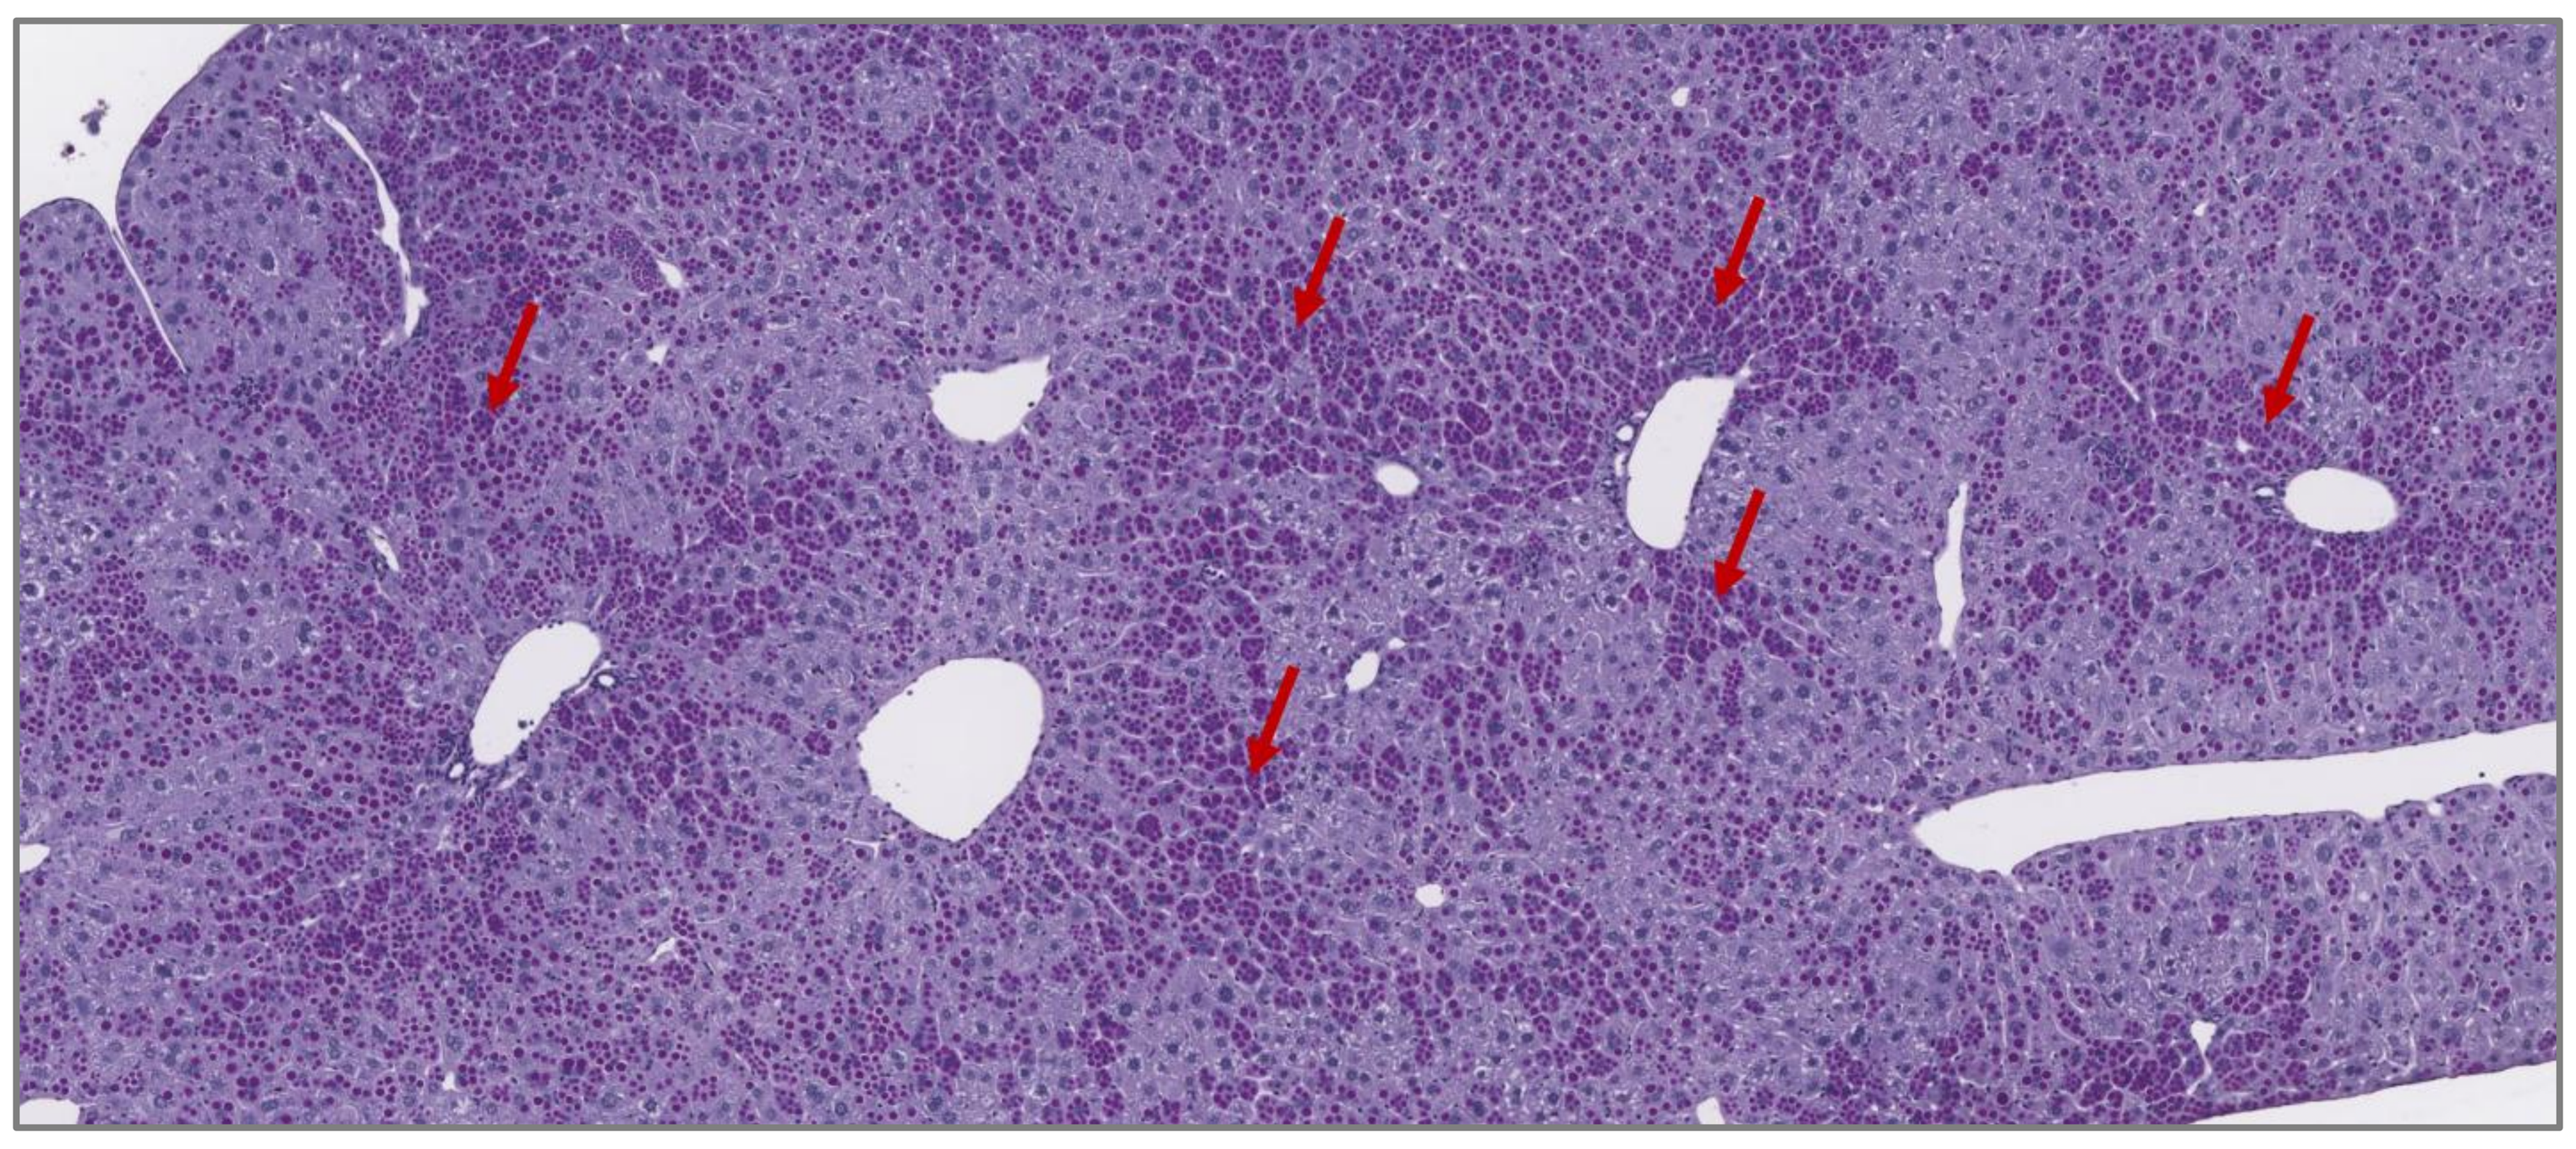

CO-8195-treated

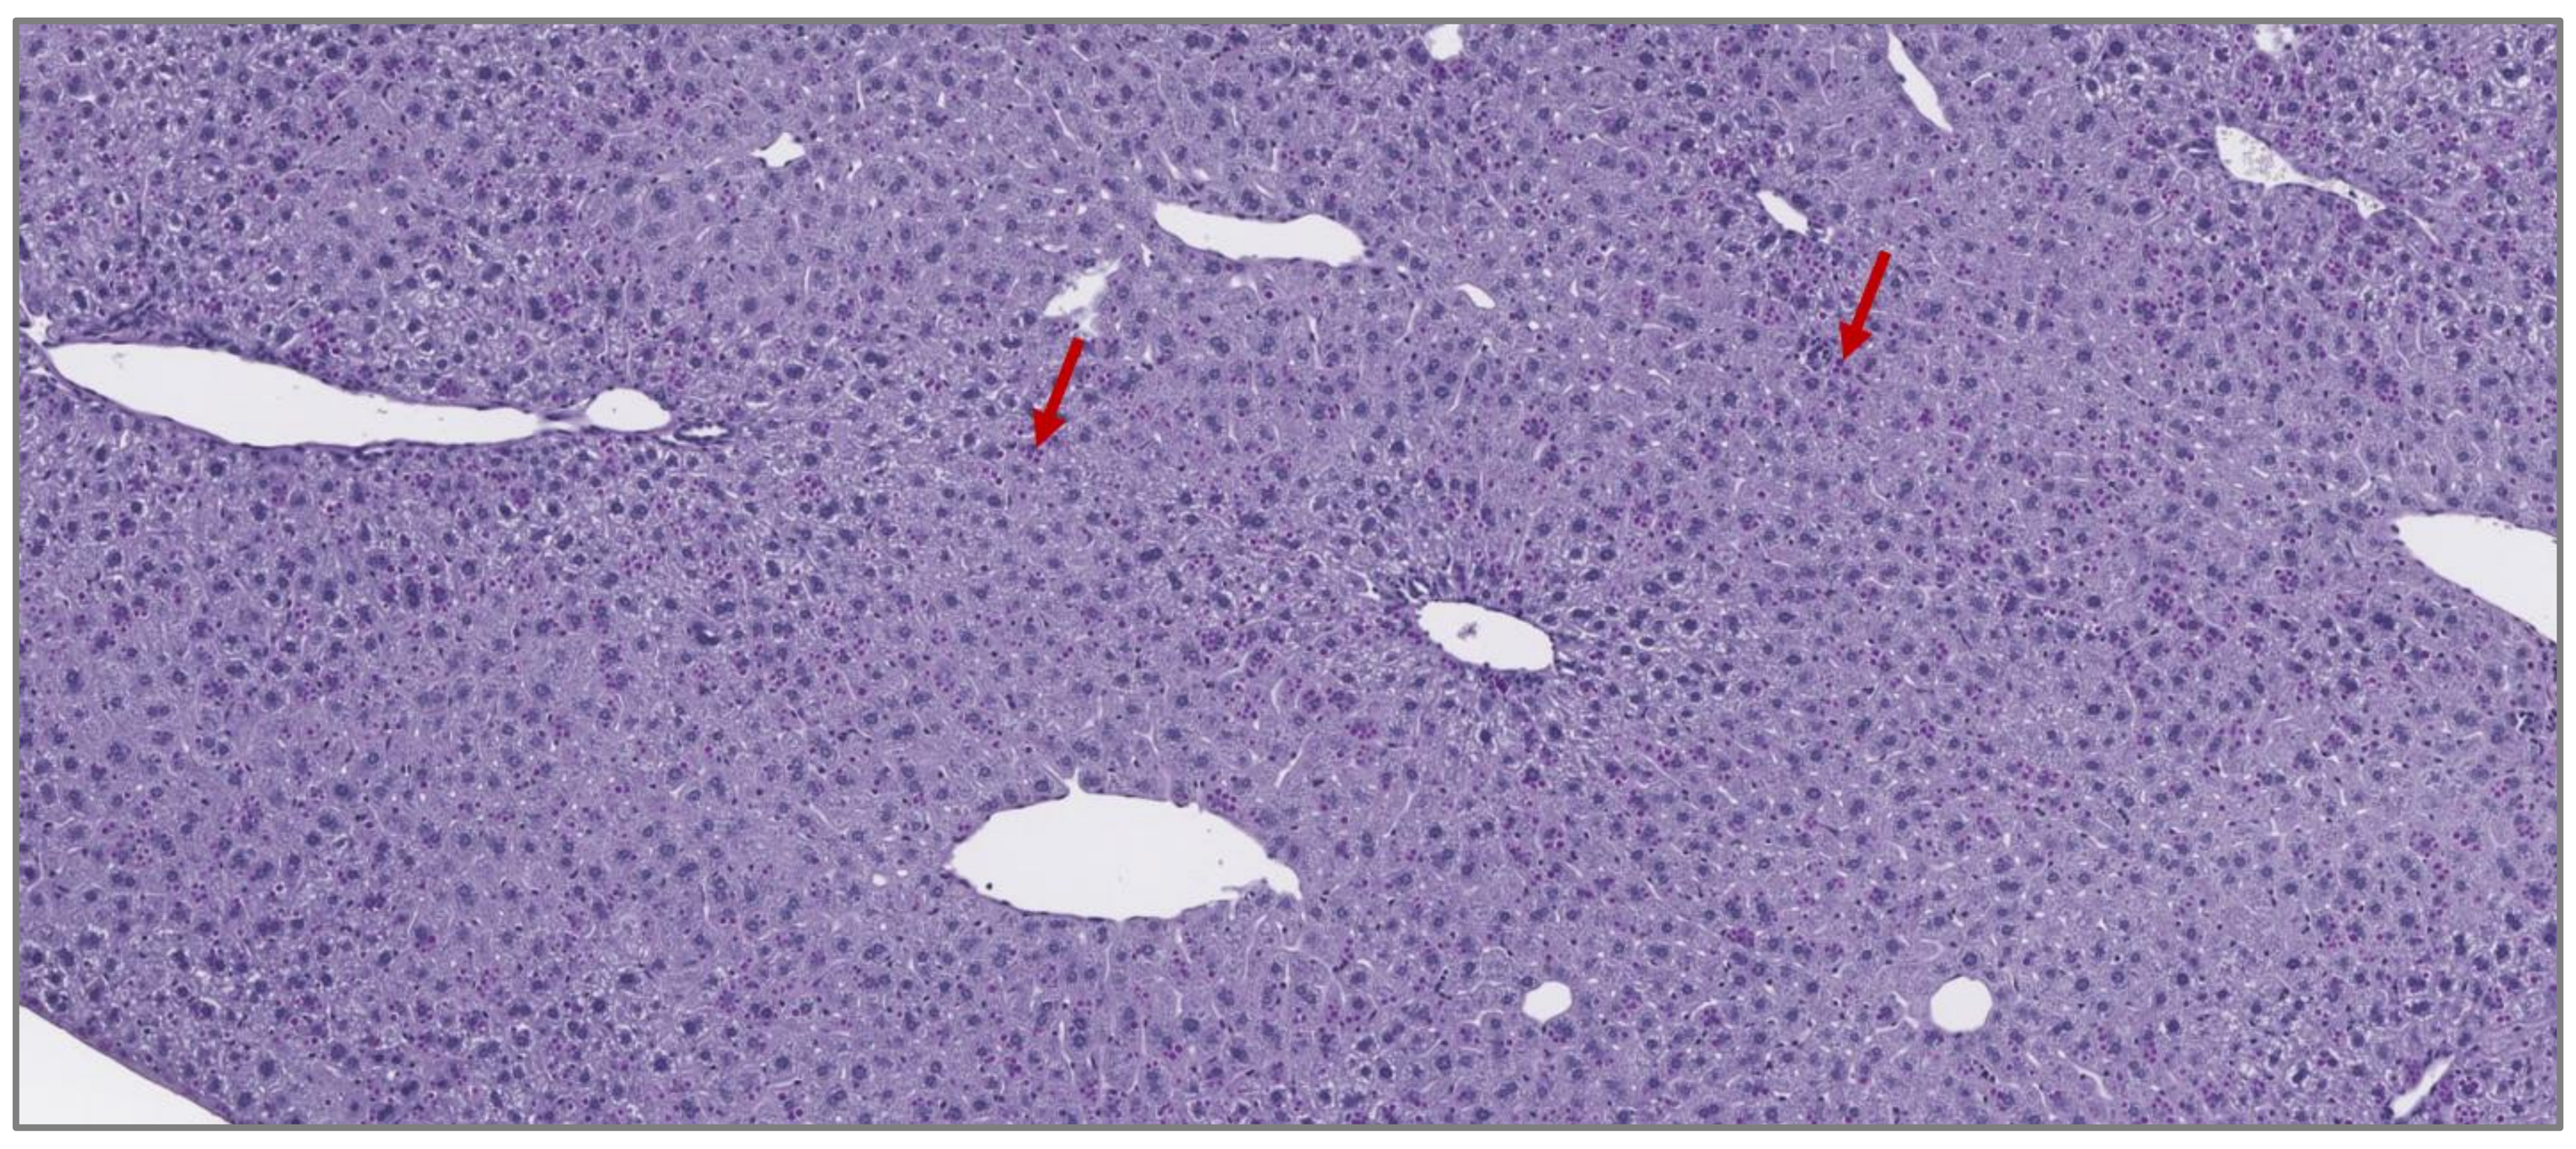

Z-A1AT polymer PAS-D (Periodic Acid-Schiff staining with diastase) staining.

CO-8195 Dose (mg/kg)	Liver polymers reduction	
	ELISA	PAS-D positivity
30	↓ 9%	↓ 43%
60	↓ 52%	↓ 59%
120	↓ 72%	↓ 72%

PAS-D positivity quantifies stained pixels in the liver section.

Conclusions

- **CO-8195 is a novel orally active small-molecule Z-A1AT corrector**
- **CO-8195 increases secretion of functional Z-A1AT and reduces Z-A1AT polymers *in vitro* and *in vivo,* supporting its potential to address both lung and liver diseases in AATD**

IND-enabling safety studies ongoing. Phase 1 initiation targeted for H1 2027

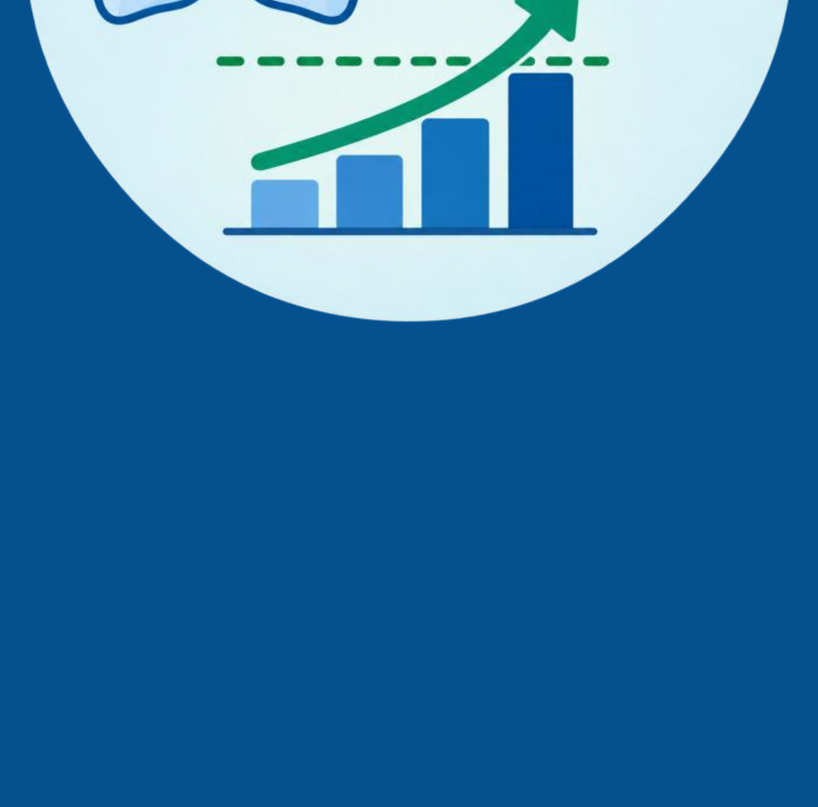

Acknowledgments: Congruence Therapeutics Team, ERS Conference Organizers

Disclosure: A.K. is an employee of Congruence Therapeutics Inc. and holds stock options

Z-A1AT, Z-alpha-1 antitrypsin; *h*NE, human neutrophile elastase; ELISA, enzyme-linked immunosorbent assay; PiZ mouse, transgenic mouse expresses human SERPINA1 (Z-A1AT)

congruence TX

Scan the QC code to find out more!